Poster #F2

Mechanism of Action of DELEXIS®, a Novel, Oral, Colonic-Targeted Drug Delivery Platform

Feng Zhang, PhD[1]; Norberto J. DeSousa, MA[2]; F. Randy Sallee, MD, PhD[3]; David Lickrish[2]; Bev Incledon, PhD[2]

[1]College of Pharmacy, Division of Molecular Pharmaceutics and Drug Delivery, University of Texas at Austin, Austin, TX, USA; [2]Ironshore Pharmaceuticals & Development, Inc., Camana Bay, Grand Cayman, Cayman Islands; [3]Ironshore Pharmaceuticals Inc., Durham, NC, USA

Introduction

- DELEXIS is a novel, oral drug delivery platform specifically engineered to address unmet needs in multiple disease states for medications that can provide targeted pan-colonic delivery, predictable time to onset of absorption, consistent level of drug release over an extended period, low peak-to-trough variability, and/or the ability to bypass the highly degradative properties of the upper gastrointestinal (GI) tract
- DELEXIS leverages several features of the GI tract to target a highly consistent delayed-release (DR) and extended-release (ER) pharmacokinetic (PK) profile
 - Size of ingested matter: Small particles (<1 mm in diameter) transit like a fluid and disperse throughout the GI tract as they transit, making them less susceptible to variations in GI transit time[1]
 - Transit time: Small intestinal transit time is constant at 3 to 4 hours, and appears to be independent of dosage form and whether one is in a fed or fasted state[2]
 - Surface area & luminal volume of the large intestine: Compared to the small intestine, the large intestine has a smaller surface area and larger luminal volume, which results in lower absorptive potential[1]
 - Shifts/Variability in pH: Mean intraluminal pH changes from an acidic environment in the stomach (pH of 1.0 to 2.5 in a fasted state up to 5.0 in a fed state), to a pH of 6.0 in the duodenum, to a highly variable pH in the ileum (7.0 to 8.0), then dropping to 5.9 in the cecum, and gradually increasing to a pH of 7.0 in the rectum[1,2]
- HLD200 is a once-daily, evening-dosed DR and ER methylphenidate (DR/ER-MPH) indicated for the treatment of attention-deficit/hyperactivity disorder (ADHD) in individuals 6 years and older. DR/ER-MPH utilizes the DELEXIS drug delivery platform to target onset of effect coinciding with awakening, continuing throughout the day, and lasting into the evening
- The PK profile of DR/ER-MPH has been evaluated in several single-center open-label studies.[3,4] The PK findings support the use of DELEXIS as a consistent and predictable drug delivery platform

Overview of the DELEXIS Drug Delivery Platform

- An overview of the DELEXIS drug delivery platform is shown in **Figure 1**

Figure 1. DELEXIS Drug Delivery Platform[1,2]

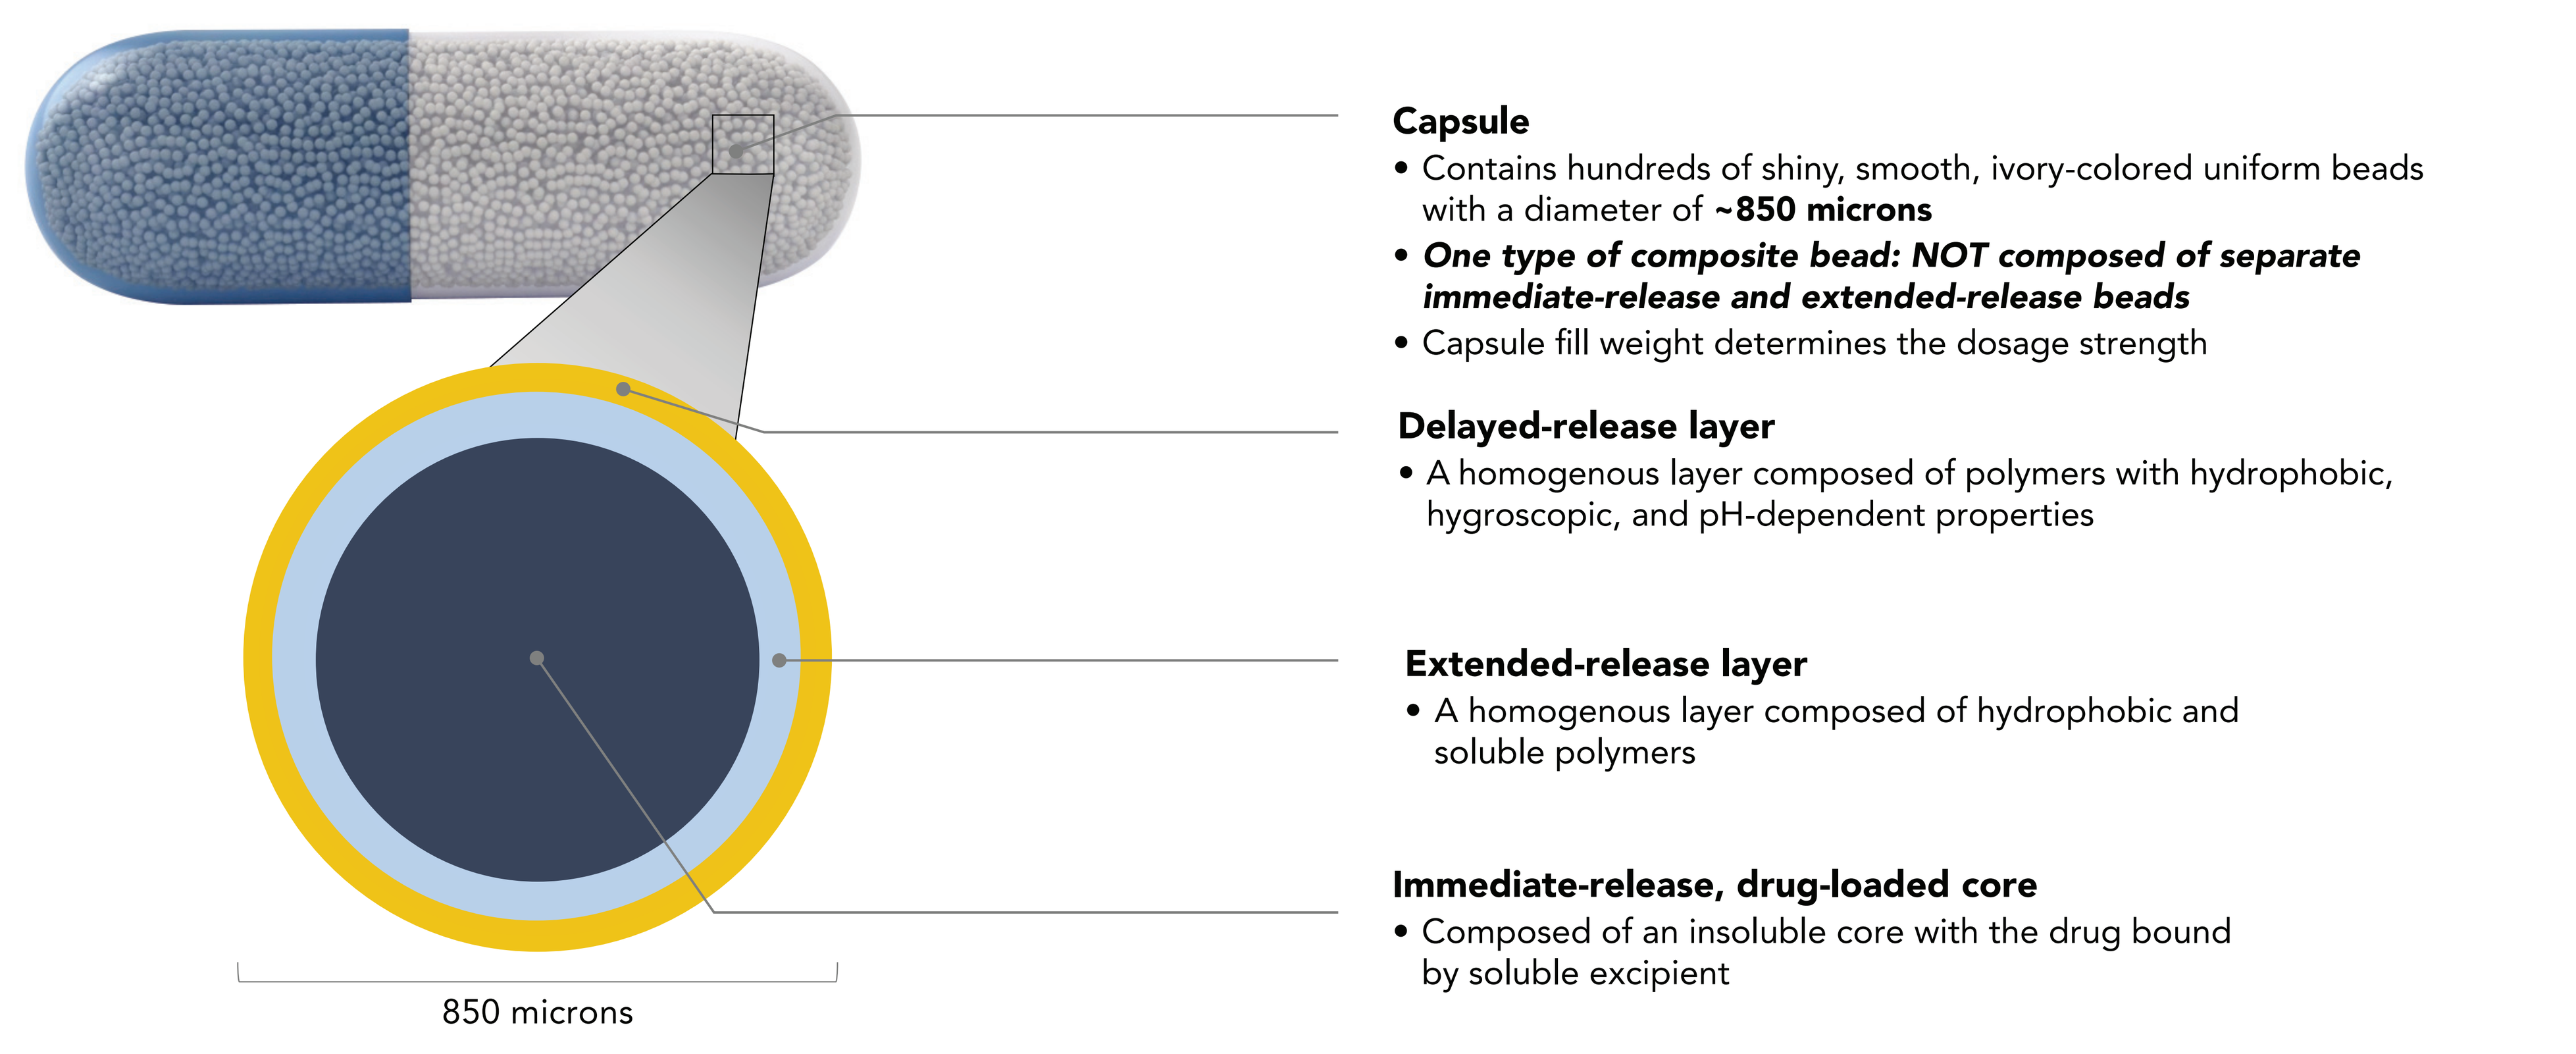
Capsule
• Contains hundreds of shiny, smooth, ivory-colored uniform beads with a diameter of ~850 microns
• One type of composite bead: NOT composed of separate immediate-release and extended-release beads
• Capsule fill weight determines the dosage strength
Delayed-release layer
• A homogenous layer composed of polymers with hydrophobic, hygroscopic, and pH-dependent properties
Extended-release layer
• A homogenous layer composed of hydrophobic and soluble polymers
Immediate-release, drug-loaded core
• Composed of an insoluble core with the drug bound by soluble excipient
850 microns

- DELEXIS utilizes proprietary microbeads (<1 mm in diameter) consisting of a drug-loaded core surrounded by two functional film layers
 - The outer DR layer has hydrophobic, hygroscopic, and pH-dependent properties, which control the time it takes for the DR layer to wet and allow fluid to access the inner ER layer
 - The ER layer has hydrophobic and soluble properties, which regulate its permeability and control dissolution from the drug-loaded core[5]
 - The drug-loaded core is composed of an insoluble microcrystalline cellulose core with the drug bound to the core by a soluble polymer
- The DR and ER layers work synergistically to provide a prolonged and predictable delay in initial drug release ≥8 hours after ingestion, followed by an extended drug release in the colon[3,5]

Mechanism of DELEXIS

- DELEXIS is designed to optimize the delivery of previously approved drug products through control of specific PK properties, including the onset and duration of drug release, site of drug absorption, and the duration of therapeutic exposure. It does this by taking advantage of timing and the physiological properties of the GI tract
- An example of how DELEXIS functions in the GI tract, in the context of DR/ER-MPH, is illustrated in **Figure 2**
- The DR layer contributes to the delay in drug absorption due to the time it takes to fully wet, which enables a prolonged and predictable delay in the onset of drug release (**Figures 2A–B**)
 - The time to wet the DR layer is controlled by the ratio of hydrophobic (water repelling) and hygroscopic (water absorbing) properties of the polymers[5]
- As the DR layer slowly begins to wet, GI fluid (ie, solvent) gains access to the pH-sensitive polymer (**Figure 2C**). Although the erosion of the DR layer does not begin until it is exposed to a pH ≥7.0, the pH threshold alone is not an independent trigger for drug release, because it is neither sufficient nor necessary for drug release
 - *In vitro* dissolution studies have shown that the DR layer does not immediately begin to dissolve at a pH of 7.4, because sufficient time for full wetting is still required
 - If a high pH environment is not met, the DR layer will still fully wet over time and allow fluid access to the inner ER layer[5-7]
- Upon wetting of the ER layer, dissolution of the soluble polymer in this layer begins (**Figure 2D**). As the ER layer becomes increasingly permeable, GI fluid gains increased access to the drug-loaded core, resulting in the release of active drug over time (**Figure 2E**)
 - The rate at which the drug diffuses out of the bead is also dependent on the drug concentration gradient between the inside and the outside of the bead[5]
- Slower absorption in the colon compared to the upper GI tract is expected to result in extended exposure following dissolution (**Figure 2F**)
- The ER layer of the DELEXIS platform does not dissolve completely because several components that compose the coating of the layer are hydrophobic and insoluble and, thus, they are excreted
- Based on the properties of the DR and ER layers, the dissolution and absorption of drug are not dependent on any single factor (eg, pH, normal variations in GI transit time, site of release), which guards against inter-patient and intra-patient variability[3,5,6,7]
- The properties of the DR and ER layers can be modified to optimize the delay before release and/or duration of controlled release for specific applications

Figure 2. DELEXIS in the Gastrointestinal Tract

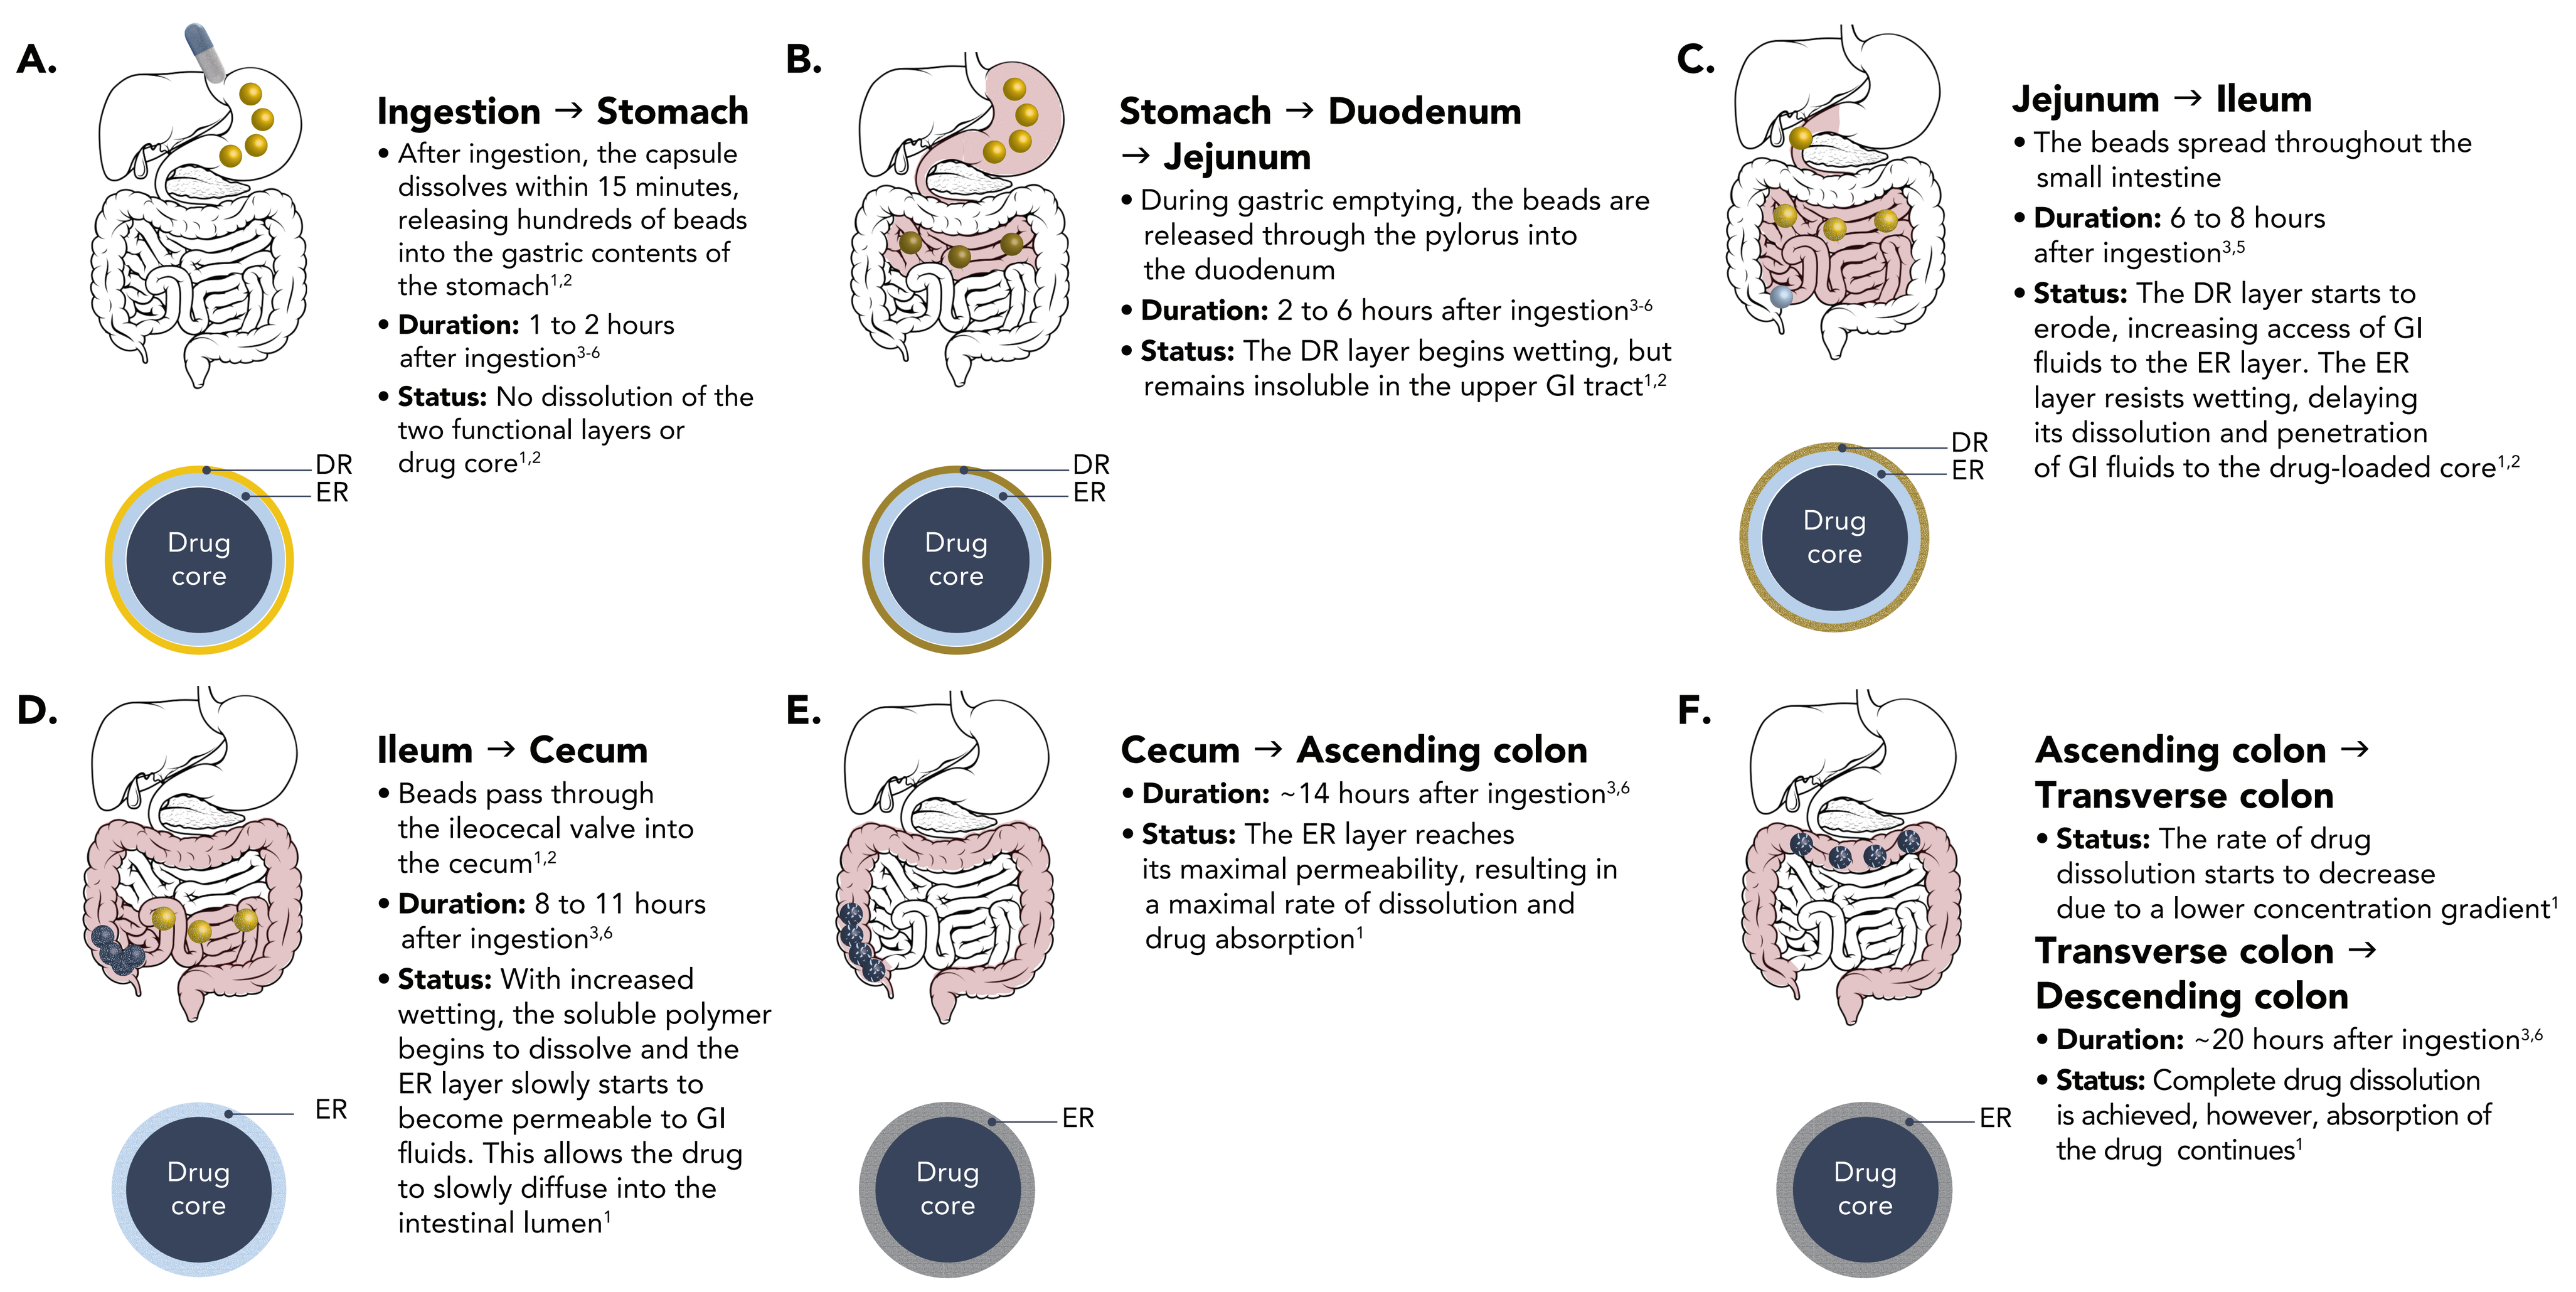
A.
Ingestion → Stomach
• After ingestion, the capsule dissolves within 15 minutes, releasing hundreds of beads into the gastric contents of the stomach[1,2]
• Duration: 1 to 2 hours after ingestion[3-6]
• Status: No dissolution of the two functional layers or drug core[1,2]
DR
ER
Drug core
B.
Stomach → Duodenum → Jejunum
• During gastric emptying, the beads are released through the pylorus into the duodenum
• Duration: 2 to 6 hours after ingestion[3-6]
• Status: The DR layer begins wetting, but remains insoluble in the upper GI tract[1,2]
DR
ER
Drug core
C.
Jejunum → Ileum
• The beads spread throughout the small intestine
• Duration: 6 to 8 hours after ingestion[3,5]
• Status: The DR layer starts to erode, increasing access of GI fluids to the ER layer. The ER layer resists wetting, delaying its dissolution and penetration of GI fluids to the drug-loaded core[1,2]
DR
ER
Drug core
D.
Ileum → Cecum
• Beads pass through the ileocecal valve into the cecum[1,2]
• Duration: 8 to 11 hours after ingestion[3,6]
• Status: With increased wetting, the soluble polymer begins to dissolve and the ER layer slowly starts to become permeable to GI fluids. This allows the drug to slowly diffuse into the intestinal lumen[1]
ER
Drug core
E.
Cecum → Ascending colon
• Duration: ~14 hours after ingestion[3,6]
• Status: The ER layer reaches its maximal permeability, resulting in a maximal rate of dissolution and drug absorption[1]
ER
Drug core
F.
Ascending colon → Transverse colon
• Status: The rate of drug dissolution starts to decrease due to a lower concentration gradient[1]
Transverse colon → Descending colon
• Duration: ~20 hours after ingestion[3,6]
• Status: Complete drug dissolution is achieved, however, absorption of the drug continues[1]
ER
Drug core

DR, delayed-release; ER, extended-release; GI, gastrointestinal.

Pharmacokinetic Profile of DR/ER-MPH Using DELEXIS

- The PK profile of DR/ER-MPH, which utilizes the DELEXIS drug delivery platform, demonstrates the DR and ER mechanism of DELEXIS (**Figure 3**)
- Following a single evening dose of DR/ER-MPH 100 mg in healthy adults (n = 20):
 - The initial absorption of MPH into plasma was delayed, and no more than 5% of the total drug was available within the first 10 hours after dosing
 - Absorption occurred in a single peak with a median T_{max} of 14.0 hours, followed by a gradual decline throughout the rest of the day
 - More than 50% of drug exposure occurred after the peak plasma concentration was reached, indicating a prolonged absorption phase[4]
- Following a single evening dose of DR/ER-MPH (54 mg) in healthy adults (n = 12) and children (n = 11) and adolescents (n = 18) with ADHD (NCT01907360), PK profiles were similar and nearly superimposable when MPH concentrations were normalized for dose and body weight across age groups[3]
- The PK profile of DR/ER-MPH is consistent with the intended mechanism of action of DELEXIS
 - Owing to its DR properties, DR/ER-MPH transits the GI tract without any overnight release of MPH, resulting in targeted delivery of MPH to the less absorptive colon
 - The protracted elimination phase supports the hypothesis that the MPH released from DR/ER-MPH is absorbed at a slower rate from the colon as opposed to the more absorptive upper GI tract, and suggests that the elimination phase consists of the sum of continued slow absorption in the colon and elimination rather than pure elimination[3]
- Individuals administered DR/ER-MPH under the same treatment condition showed low inter-subject variability in the time to achieve T_{max} (coefficient of variation [CV] ≤14.2%)[3,4]
- Low intra-subject variability was also reported for total MPH exposure (CV = 12.4%) and peak MPH concentration (CV = 20.1%; **Table 1**)[4]

Figure 3. Mean Methylphenidate Concentrations Over Time After a Single Dose of DR/ER-MPH[4,a]

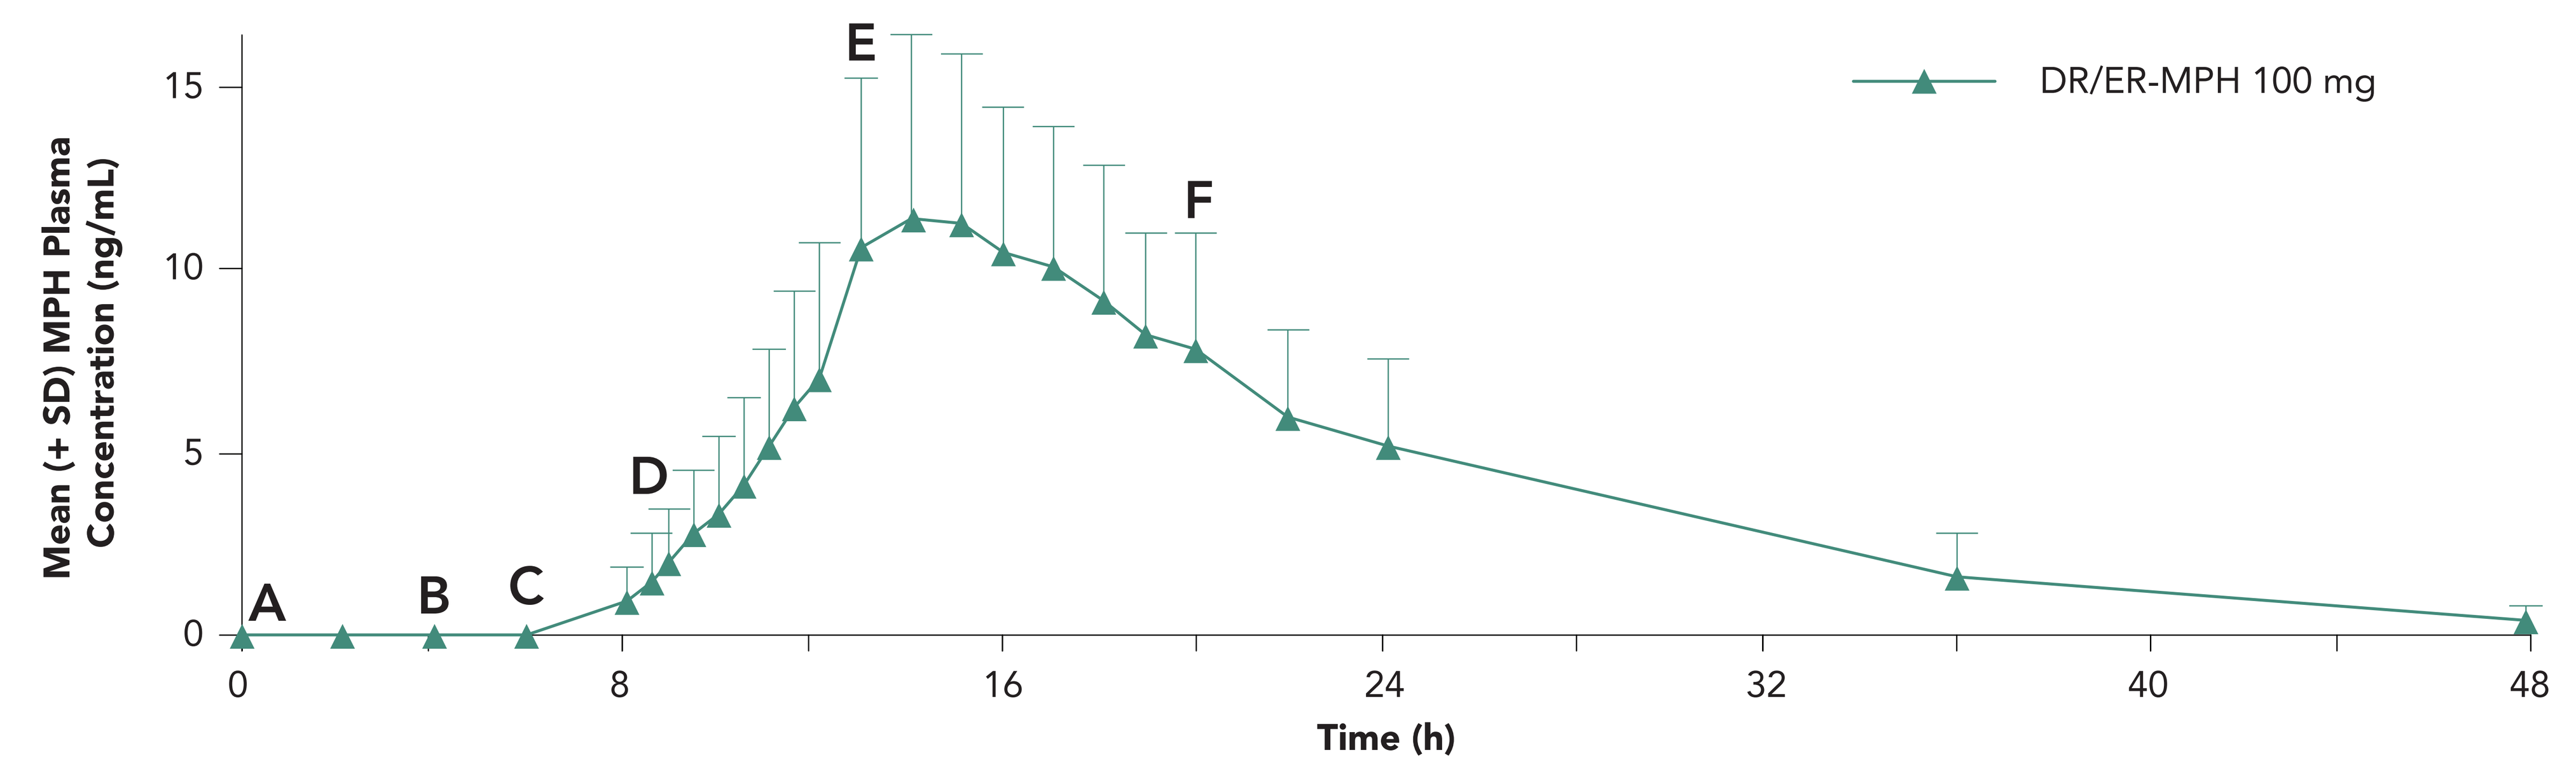
DR/ER-MPH 100 mg
Mean (+ SD) MPH Plasma Concentration (ng/mL)
15
10
5
0
A
B
C
D
E
F
0
8
16
24
32
40
48
Time (h)

[a]The letters on the graph correspond to the panels in Figure 2.
DR/ER-MPH, delayed-release and extended-release methylphenidate; MPH, methylphenidate; SD, standard deviation.

Table 1. Low Intra-Subject Variability in Key PK Parameters of DR/ER-MPH[4]

Parameter	Intra-subject CV (%)
Dose-normalized AUC_{0-t} ([ng•hr/mL]/mg)	12.42
Dose-normalized C_{max} ([ng/mL]/mg)	20.06

AUC_{0-t}, area under the time-concentration curve from time zero (pre-dose) to time of last quantifiable concentration; C_{max}, peak plasma concentration; CV, coefficient of variation; DR/ER-MPH, delayed-release and extended-release methylphenidate; PK, pharmacokinetic.

Conclusions

- DELEXIS, a novel, oral, DR and ER drug delivery platform, leverages the properties of the GI tract to achieve a unique pharmacokinetic profile
- By modifying the DR and ER layers, DELEXIS can be adapted for a number of conditions – to target precise onset and duration of drug release, site of drug absorption, and duration of effect
- The PK profile of DR/ER-MPH supports the use of DELEXIS as a colonic-targeted DR and ER drug delivery platform that provides a consistent time to onset of drug absorption and a prolonged drug release over an extended period

Disclosures

Feng Zhang, PhD: Patent-holder – Ironshore Pharmaceuticals & Development, Inc.
Norberto J. DeSousa, MA: Employee – Ironshore Pharmaceuticals & Development, Inc.
F. Randy Sallee, MD, PhD: Employee – Ironshore Pharmaceuticals Inc.; Advisory Board/Board of Directors – P2D Bioscience
David Lickrish: Employee, Patent-holder – Ironshore Pharmaceuticals & Development, Inc.
Bev Incledon, PhD: Employee – Ironshore Pharmaceuticals & Development, Inc.

References

1. Bowles A, et al. *Int J Pharm*. 2010;395:37-43. **2.** Watts P, Illum L. *Drug Dev Ind Pharm*. 1997;23(9):893-913. **3.** Childress AC, et al. *J Child Adolesc Psychopharmacol*. 2018;28(1):10-18. **4.** Liu T, et al. *J Child Adolesc Psychopharmacol*. 2019; 29(3):1-11. **5.** Ironshore Pharmaceuticals & Development, Inc. Data on file: Patent # 8,927,010 (Compositions MPH). **6.** Ibekwe VC, et al. *J Pharm Sci*. 2006;95(12):2760-2766. **7.** Watts P, et al. *Int J Pharm*. 1992;87:215-221.

Acknowledgements

Editorial assistance was provided by inScience Communications, Springer Healthcare (Philadelphia, PA, USA).

Supported by funding from Ironshore Pharmaceuticals & Development, Inc.

Poster presented at the American Professional Society of ADHD and Related Disorders (APSARD) Annual Meeting, January 17–19, 2020.